In adult macaque monkeys subjected to an incomplete spinal cord injury, numerous large SMI-32 positive fibers penetrate the scar tissue

M.-L. Beaud[1], P. Freund[1], E. Schmidlin[1], J. Bloch[3], A. Mir[4], M.E. Schwab[2], E.M. Rouiller[1] and T. Wannier[1,2].

1: Dept of Medicine, Uni Fribourg; 2: Brain Research Inst., Uni Zurich; 3: CHUV, Lausanne; 4: Novartis, Basel

Grant Sponsors: *Swiss National Science Foundation, grants No 31-43422.95, 4038-43918, 31-61857.00, 31-120005 (EMR); Novartis Foundation; The National Centre of Competence in Research (NCCR) on "Neural plasticity", Ch. Reeves Foundation*

INTRODUCTION

• Following an incomplete cervical cord hemisection (level C7/C8), corticospinal (CS) fibers of adult macaque monkeys treated with an anti-Nogo-A antibody sprout rostrally and caudally to the lesion site. However, as assessed using the anterograde tracer BDA injected in motor cortex, only few CS axons were found to penetrate into the scar tissue (Freund et al., 2006).

• As different types of neurons respond differently to injury, the question arises whether some axons, different from CS axons, penetrate and eventually cross the scar tissue.

• To investigate this question, we used the SMI-32 antibody recognizing fibers containing neurofilaments inside the scar tissue. Furthermore, we investigated whether the presence of growth promoting molecules had an impact on the number and/or the cumulated lenght of SMI-32 positive (+) fibers.

MATERIALS & METHODS

Sixteen young adult macaque monkeys (♂) were subjected to an incomplete unilateral section of the spinal cord at the C7/C8 level. The animals were treated with either a control antibody (n=8), or an anti-Nogo-A antibody (n=5), or an anti-Nogo-A antibody combined with BNDF (n=3).
We investigated the number and the cumulative length of SMI-32 (+) fibers present into the lesion site of each groups of animals.

CONCLUSIONS

Conclusion 1
After a spinal cord lesion, the scar tissue can be successfully colonized by certain axons.

Conclusion 2
Ventral root axons penetrate the lesion, and numerous fibers observed into the scar tissue have a similar size and quality of staining as ventral root axons.
We therefore propose that most of these fibers originate from motoneurons.

Conclusion 3
Neither the anti-Nogo-A antibody alone, nor the combination of this antibody with BDNF influences the number or the length of fibers inside the scar tissue.

RESULTS

1) Reconstruction and extent of the spinal cord lesion

The extent of the incomplete spinal hemisection varied among animals, ranging between 38 % and 95 %.
Numerous descending & ascending tracts were interrupted ipsilesionally.

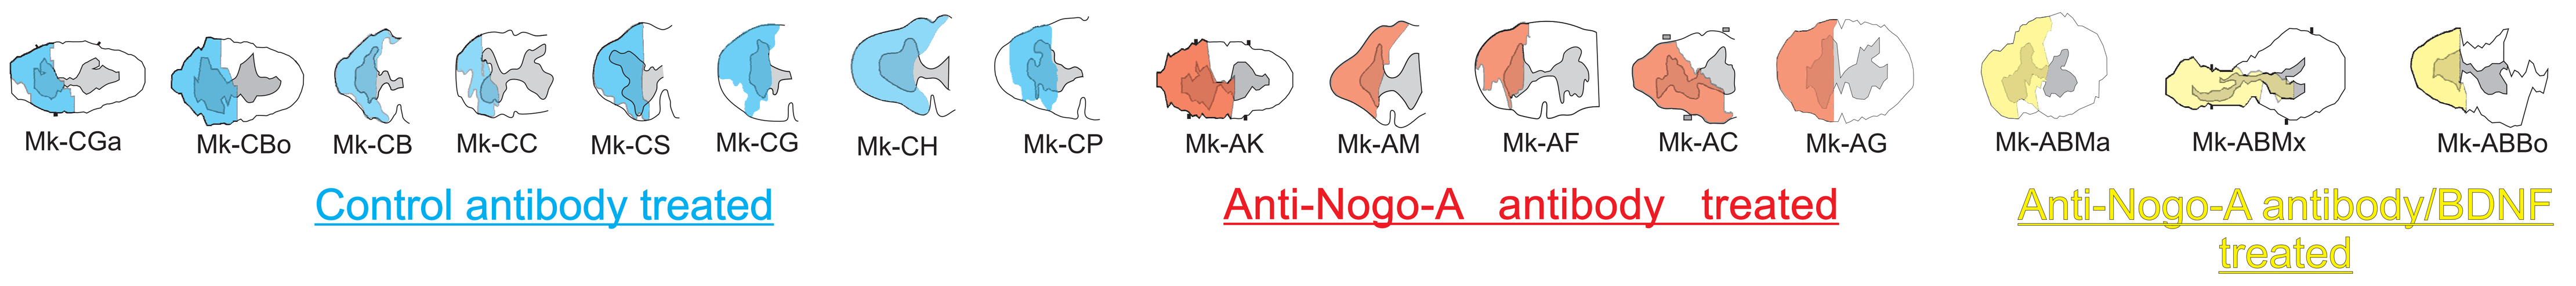

Mk-CGa
Mk-CBo
Mk-CB
Mk-CC
Mk-CS
Mk-CG
Mk-CH
Mk-CP
Mk-AK
Mk-AM
Mk-AF
Mk-AC
Mk-AG
Mk-ABMa
Mk-ABMx
Mk-ABBo
Control antibody treated
Anti-Nogo-A antibody treated
Anti-Nogo-A antibody/BDNF treated

2) Which spinal elements are labelled by the SMI-32 staining?

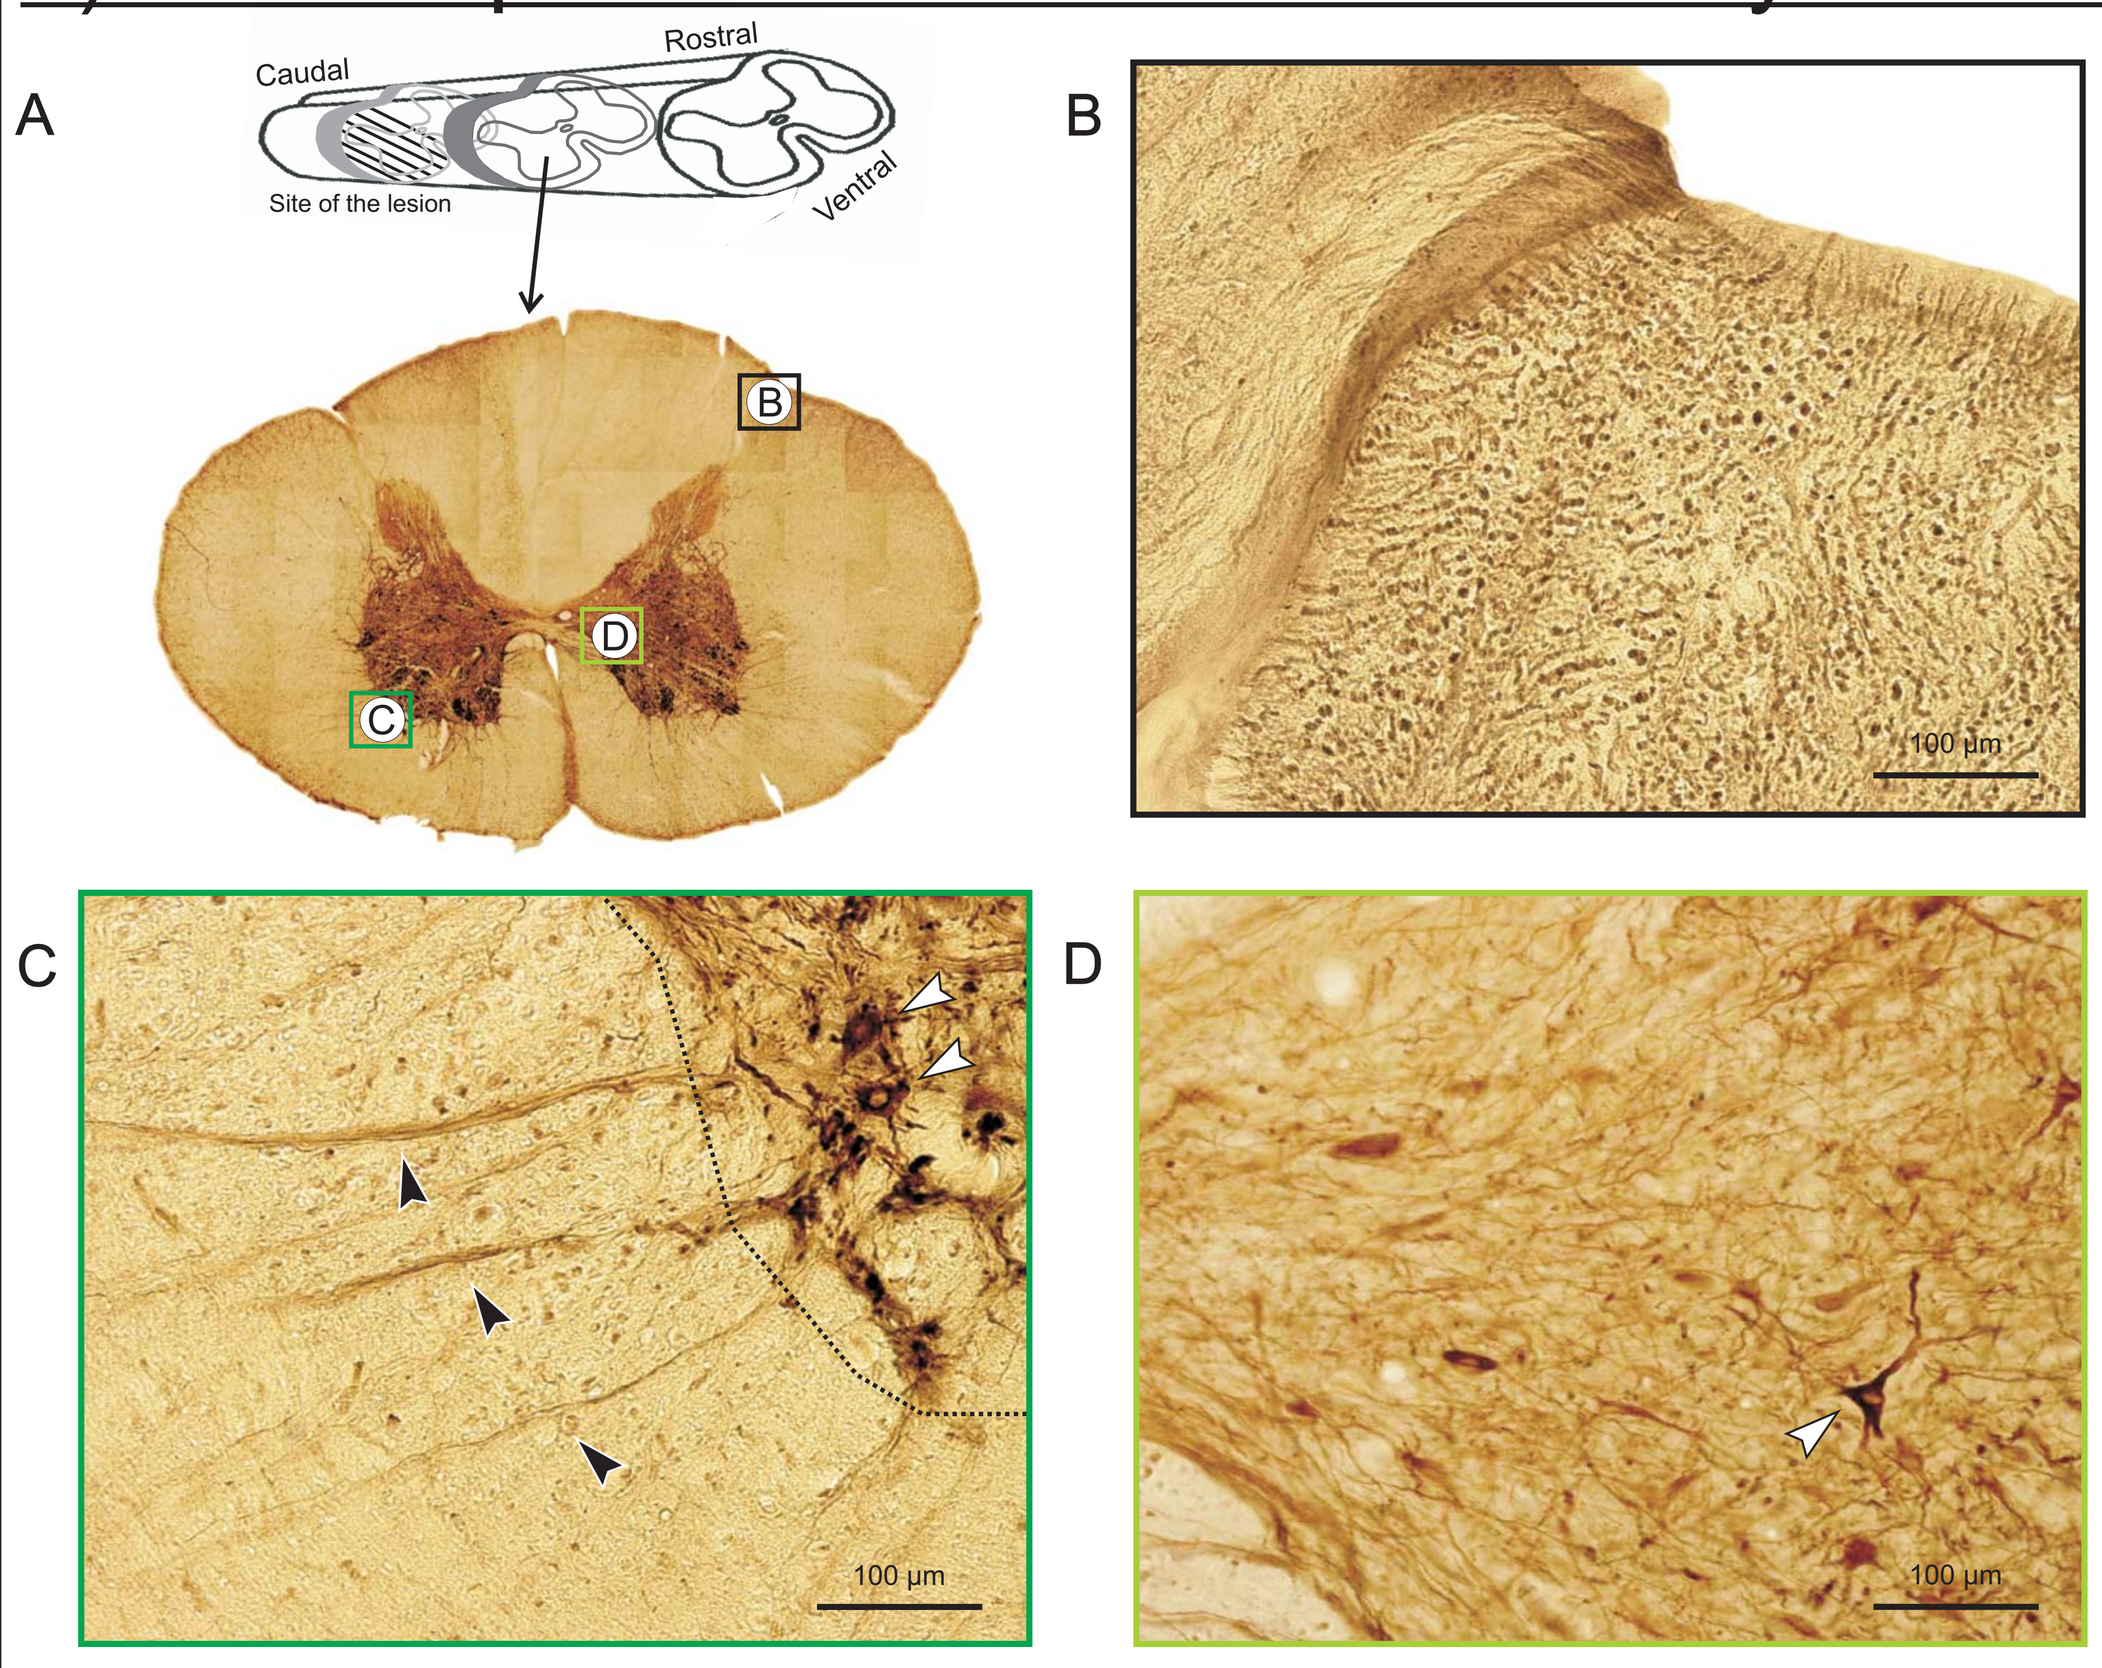

A
Caudal
Rostral
Site of the lesion
Ventral
B
C
D
100 µm

•Among spinal neurons, motoneurons were the most heavily SMI-32 stained neurons (C, white arrowheads).
•Few additional heavily stained neurons were scattered inside the entire gray matter (D, white arrowheads).

•In addition to neurons, fibers such as the CS fibers (B) or motoneurons projections (C, black arrowheads) were also distinctly stained.

3) The SMI-32 staining reveals numerous fibers inside the scar tissue; most were located ventrally.

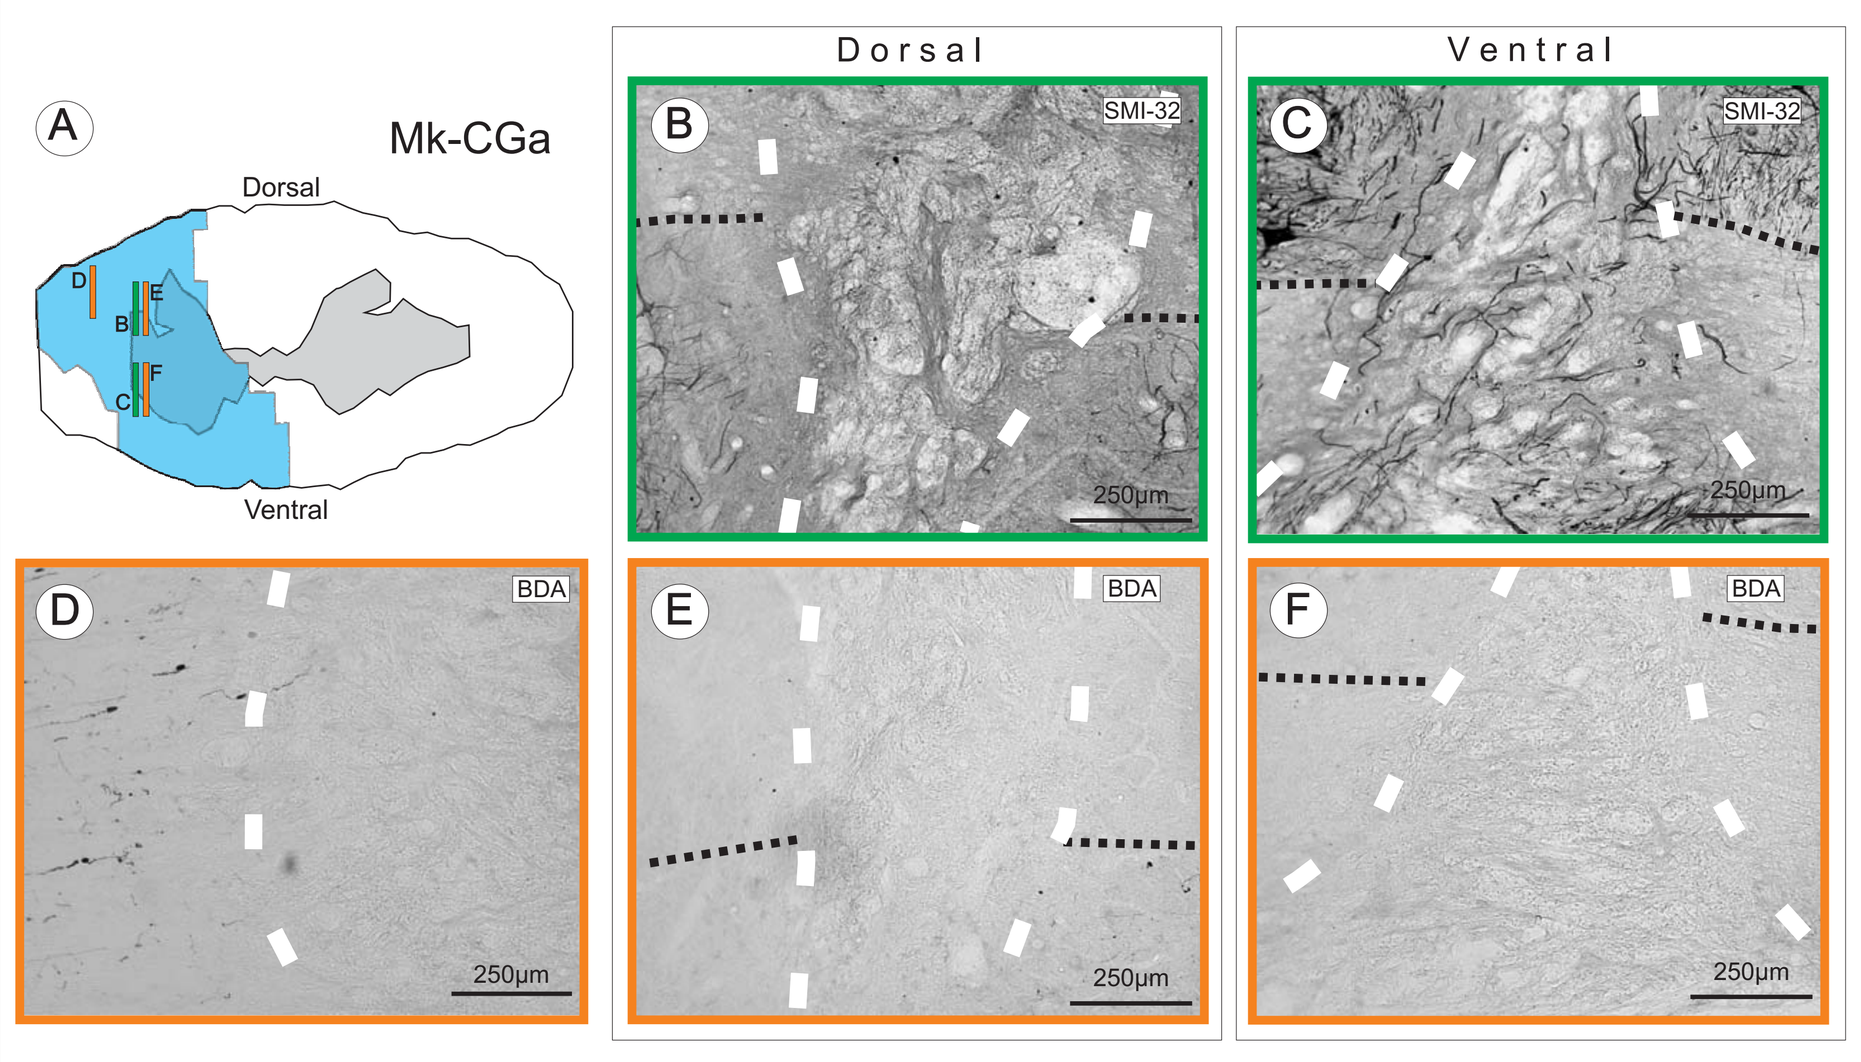

A
Mk-CGa
Dorsal
Ventral
Dorsal
Ventral
B
C
SMI-32
D
E
F
BDA
250µm

• Regardless of the dorso-ventral position, no BDA (+) fibers are visible in the scar tissue (E,F).
• Also in dorsolateral funiculus location, few BDA (+) CS fibers attempt to penetrate the scar tissue (D).

• The SMI-32 staining reveals numerous (+) fibers especially in the ventral part of the scar tissue (B, C).

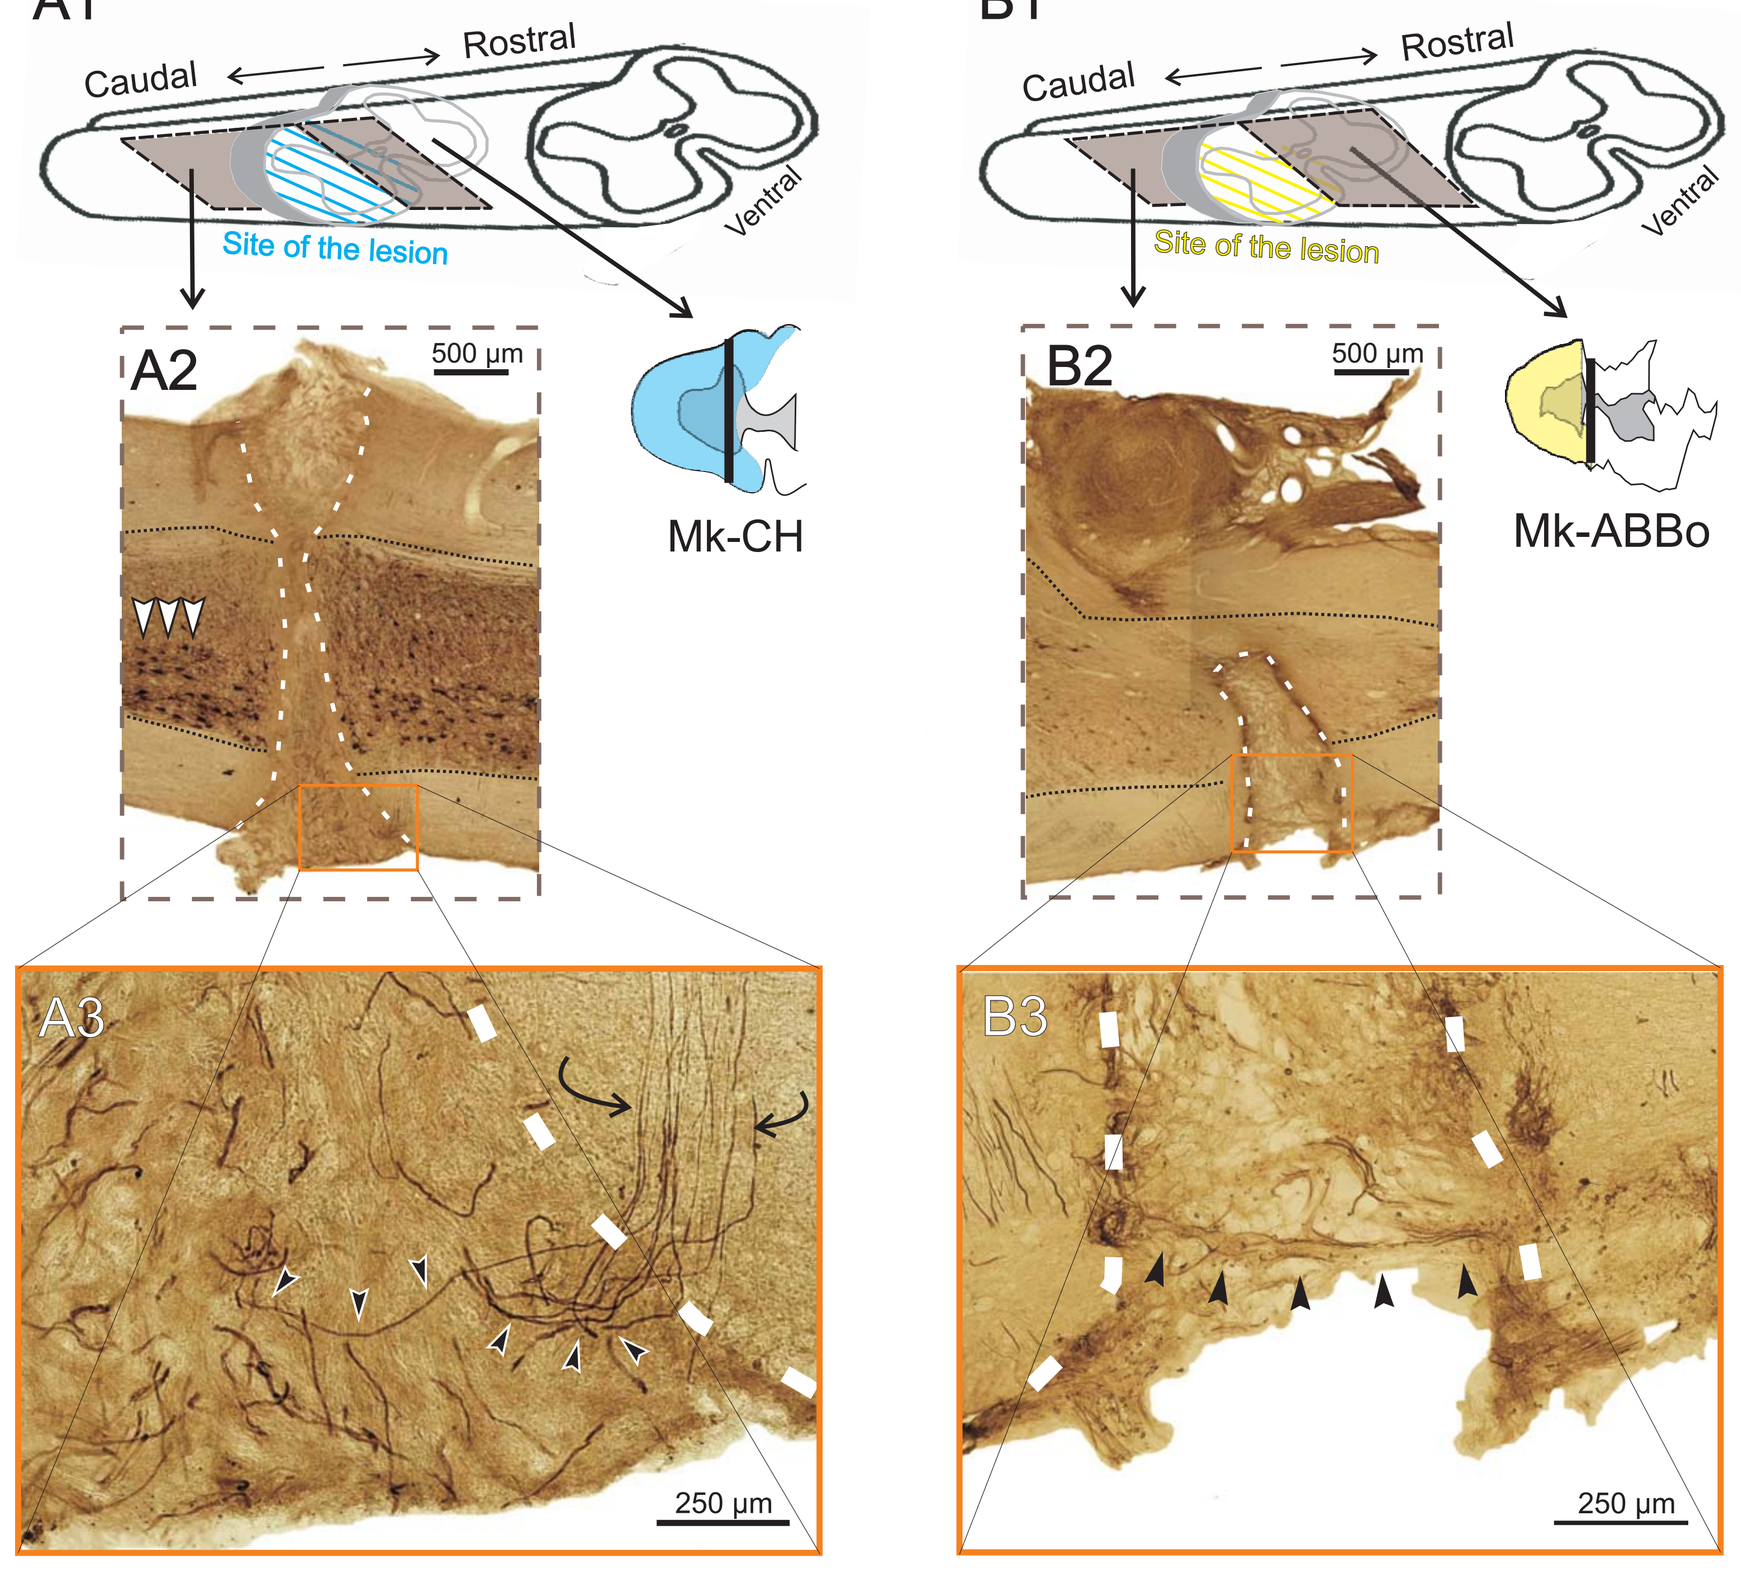

A1
Caudal
Rostral
Site of the lesion
Ventral
A2
500 µm
Mk-CH
A3
250 µm
B1
B2
Mk-ABBo
B3

•Numerous SMI-32 (+) fibers invade the scar tissue (A,B). Most are irregularly oriented (A3) but some cross the rostro-caudal axis of the lesion (B3).

•Some of those axons, have a segment inside the intact white matter, where they are included in bundles otherwise forming ventral roots (A3, curved arrows). This demonstrates that some of these fibers belong to motoneurons.

4) The scar tissue contains SMI-32 (+) fibers of different diameter.

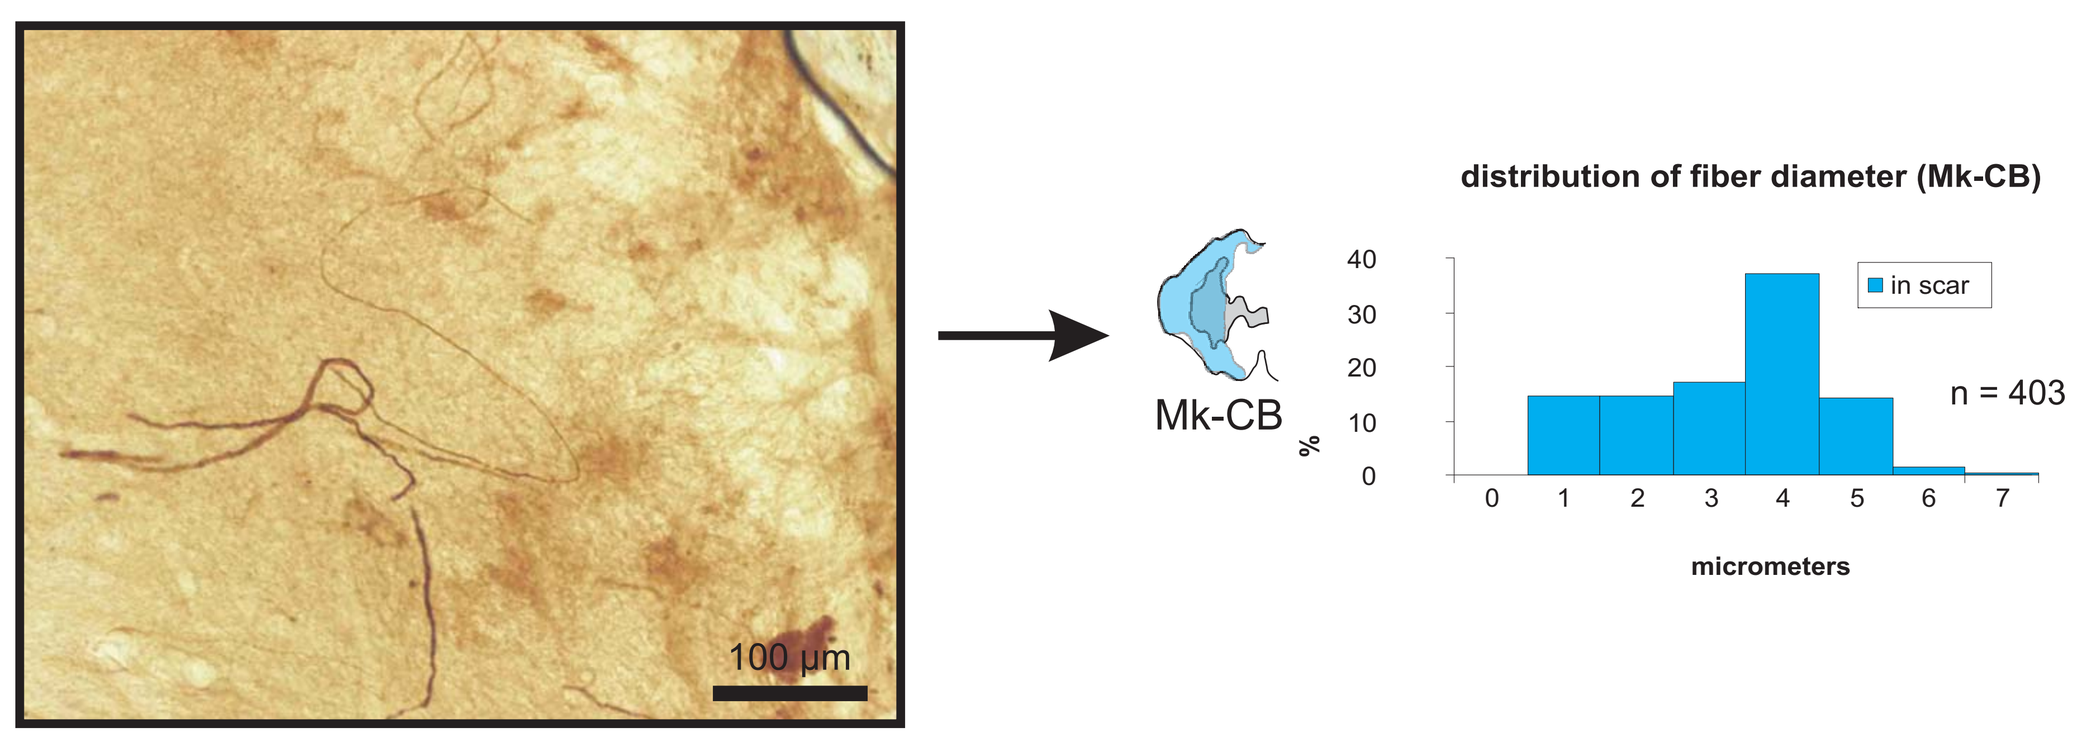

100 µm
Mk-CB
distribution of fiber diameter (Mk-CB)
in scar
n = 403
micrometers

• The diameter of SMI-32 (+) fibers observed into the scar ranges from thin (< 2 µm) to large fibers (> 6 µm).

5) The distribution of SMI-32 (+) fibers diameter in the scar and in the ventral white matter overlap but differ.

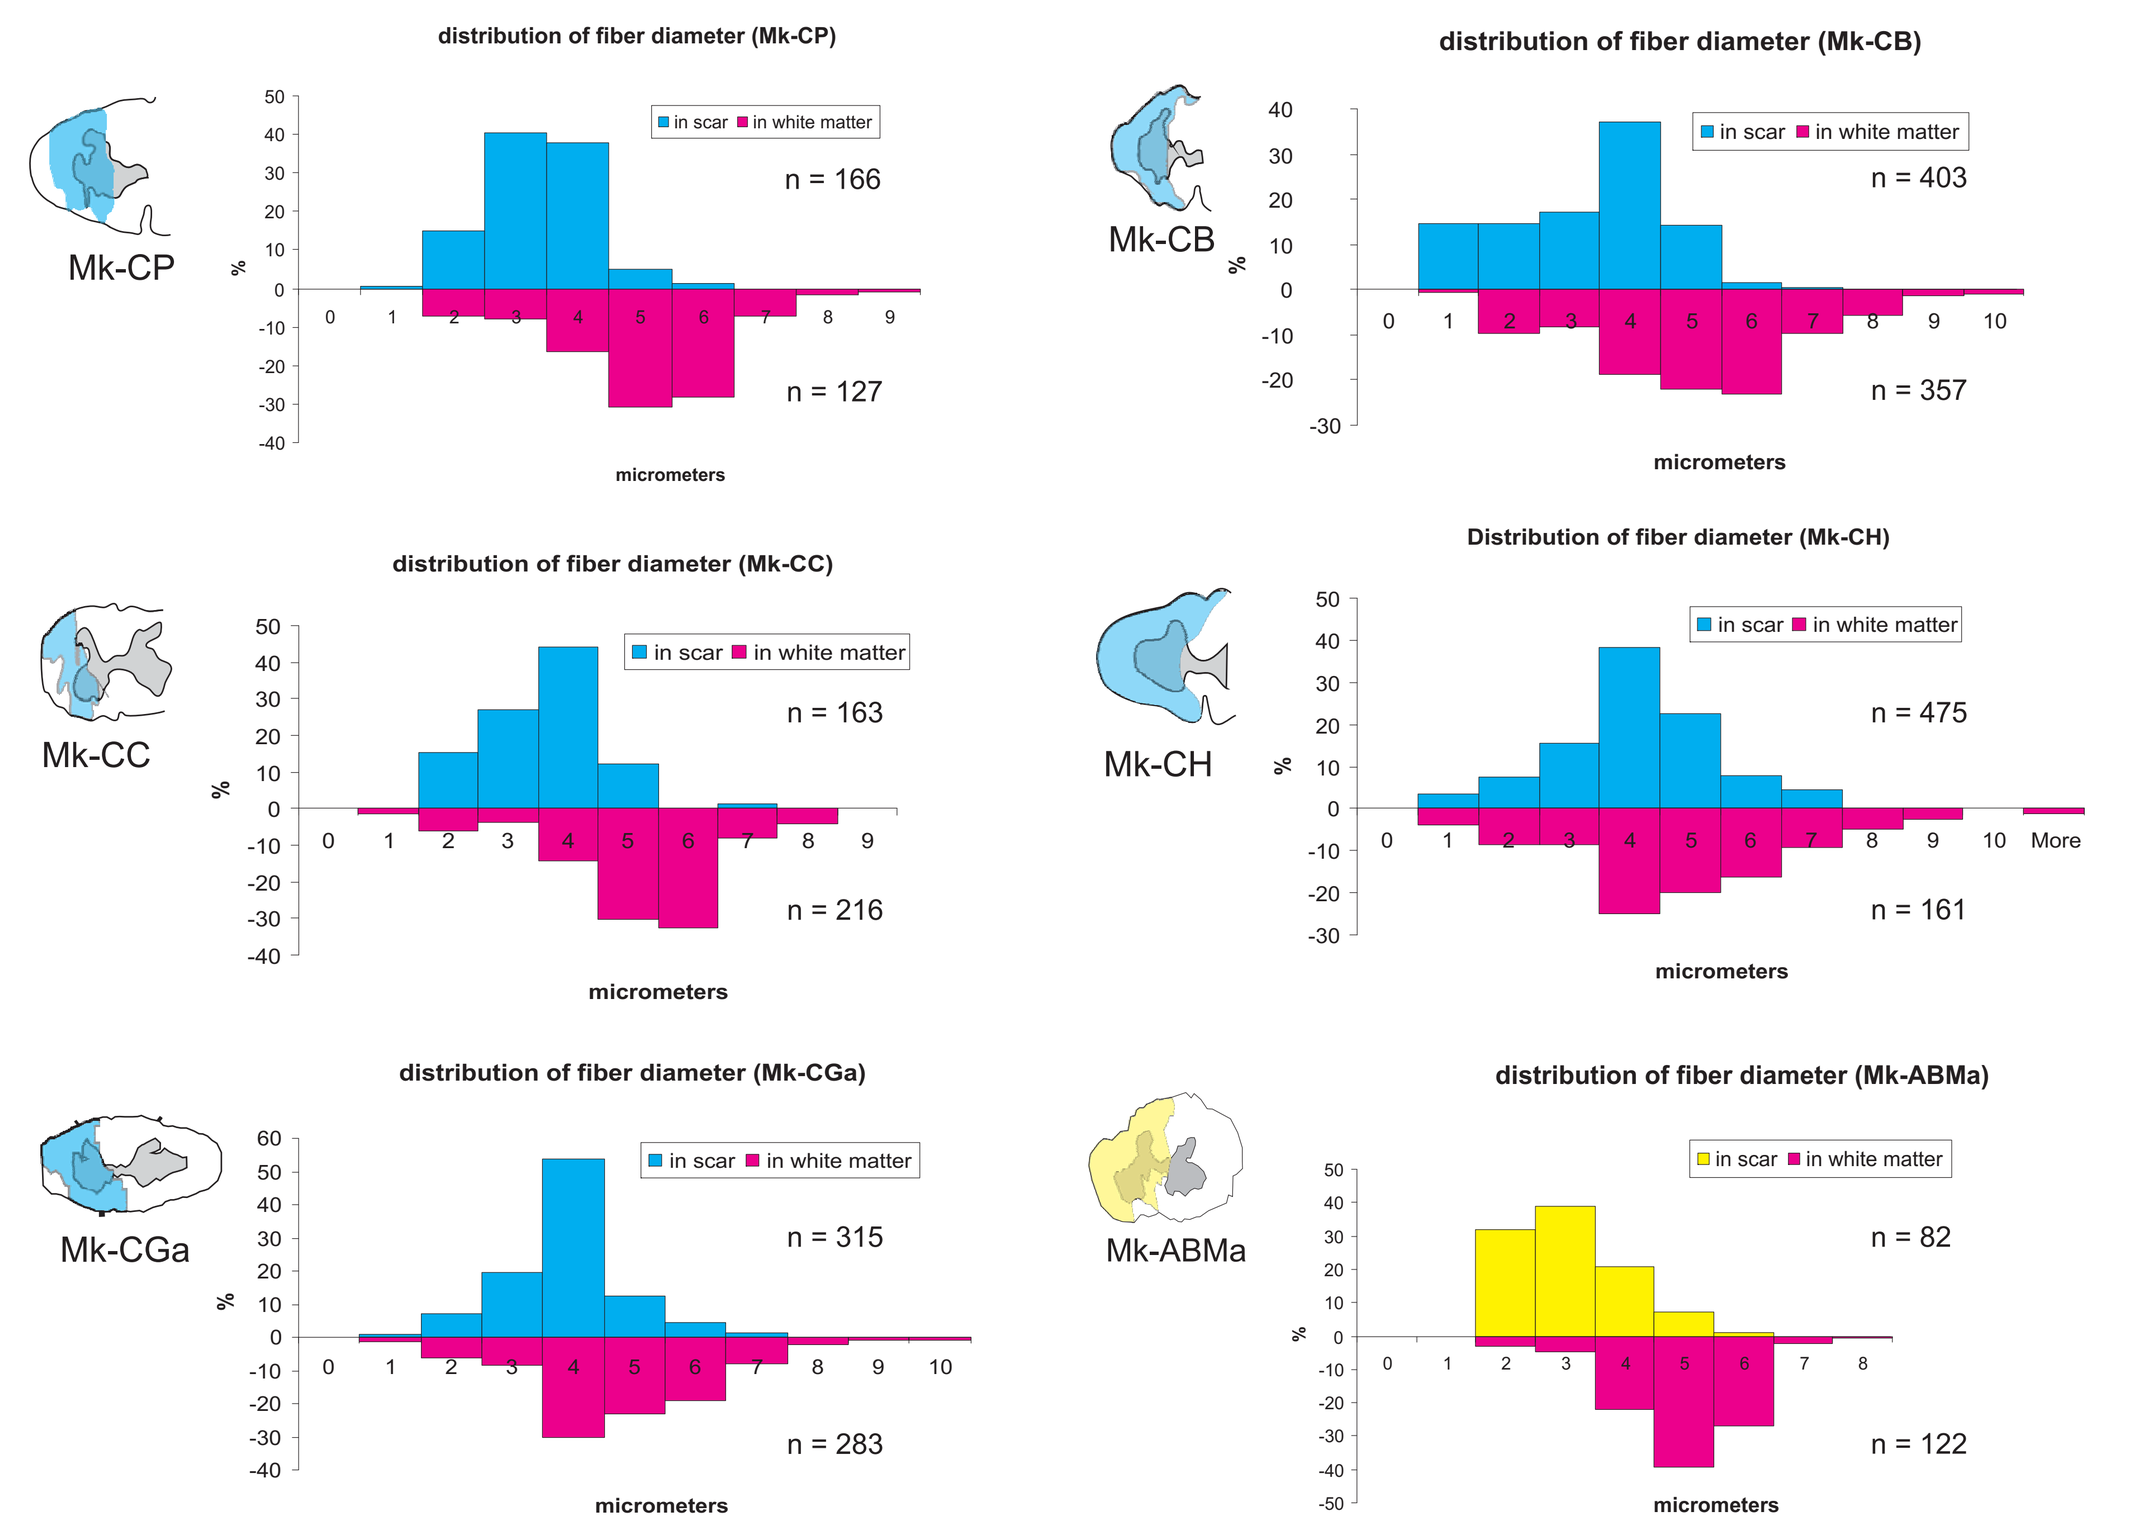

distribution of fiber diameter (Mk-CP)
Mk-CP
n = 166
n = 127
distribution of fiber diameter (Mk-CB)
Mk-CB
n = 403
n = 357
distribution of fiber diameter (Mk-CC)
Mk-CC
n = 163
n = 216
Distribution of fiber diameter (Mk-CH)
Mk-CH
n = 475
n = 161
distribution of fiber diameter (Mk-CGa)
Mk-CGa
n = 315
n = 283
distribution of fiber diameter (Mk-ABMa)
Mk-ABMa
n = 82
n = 122
in scar
in white matter
micrometers

• This indicates that fibers from neurons other than motoneurons invade also the scar.

• *In oder to evaluate whether different types of axons responded differently to the presence of growth promoting molecules, we decided to split arbitrarily these fibers populations into two groups:*
- *a "thick fibers" group (∅ > 3.1 µm), presumably containing a high proportion of motoneurons axons.*
- *a "thin fibers" group (∅ < 3.1 µm), presumably containing a large proportion of non-motoneuron axons.*

6) Some of the "thin" fibers are not coming from the ventral roots.

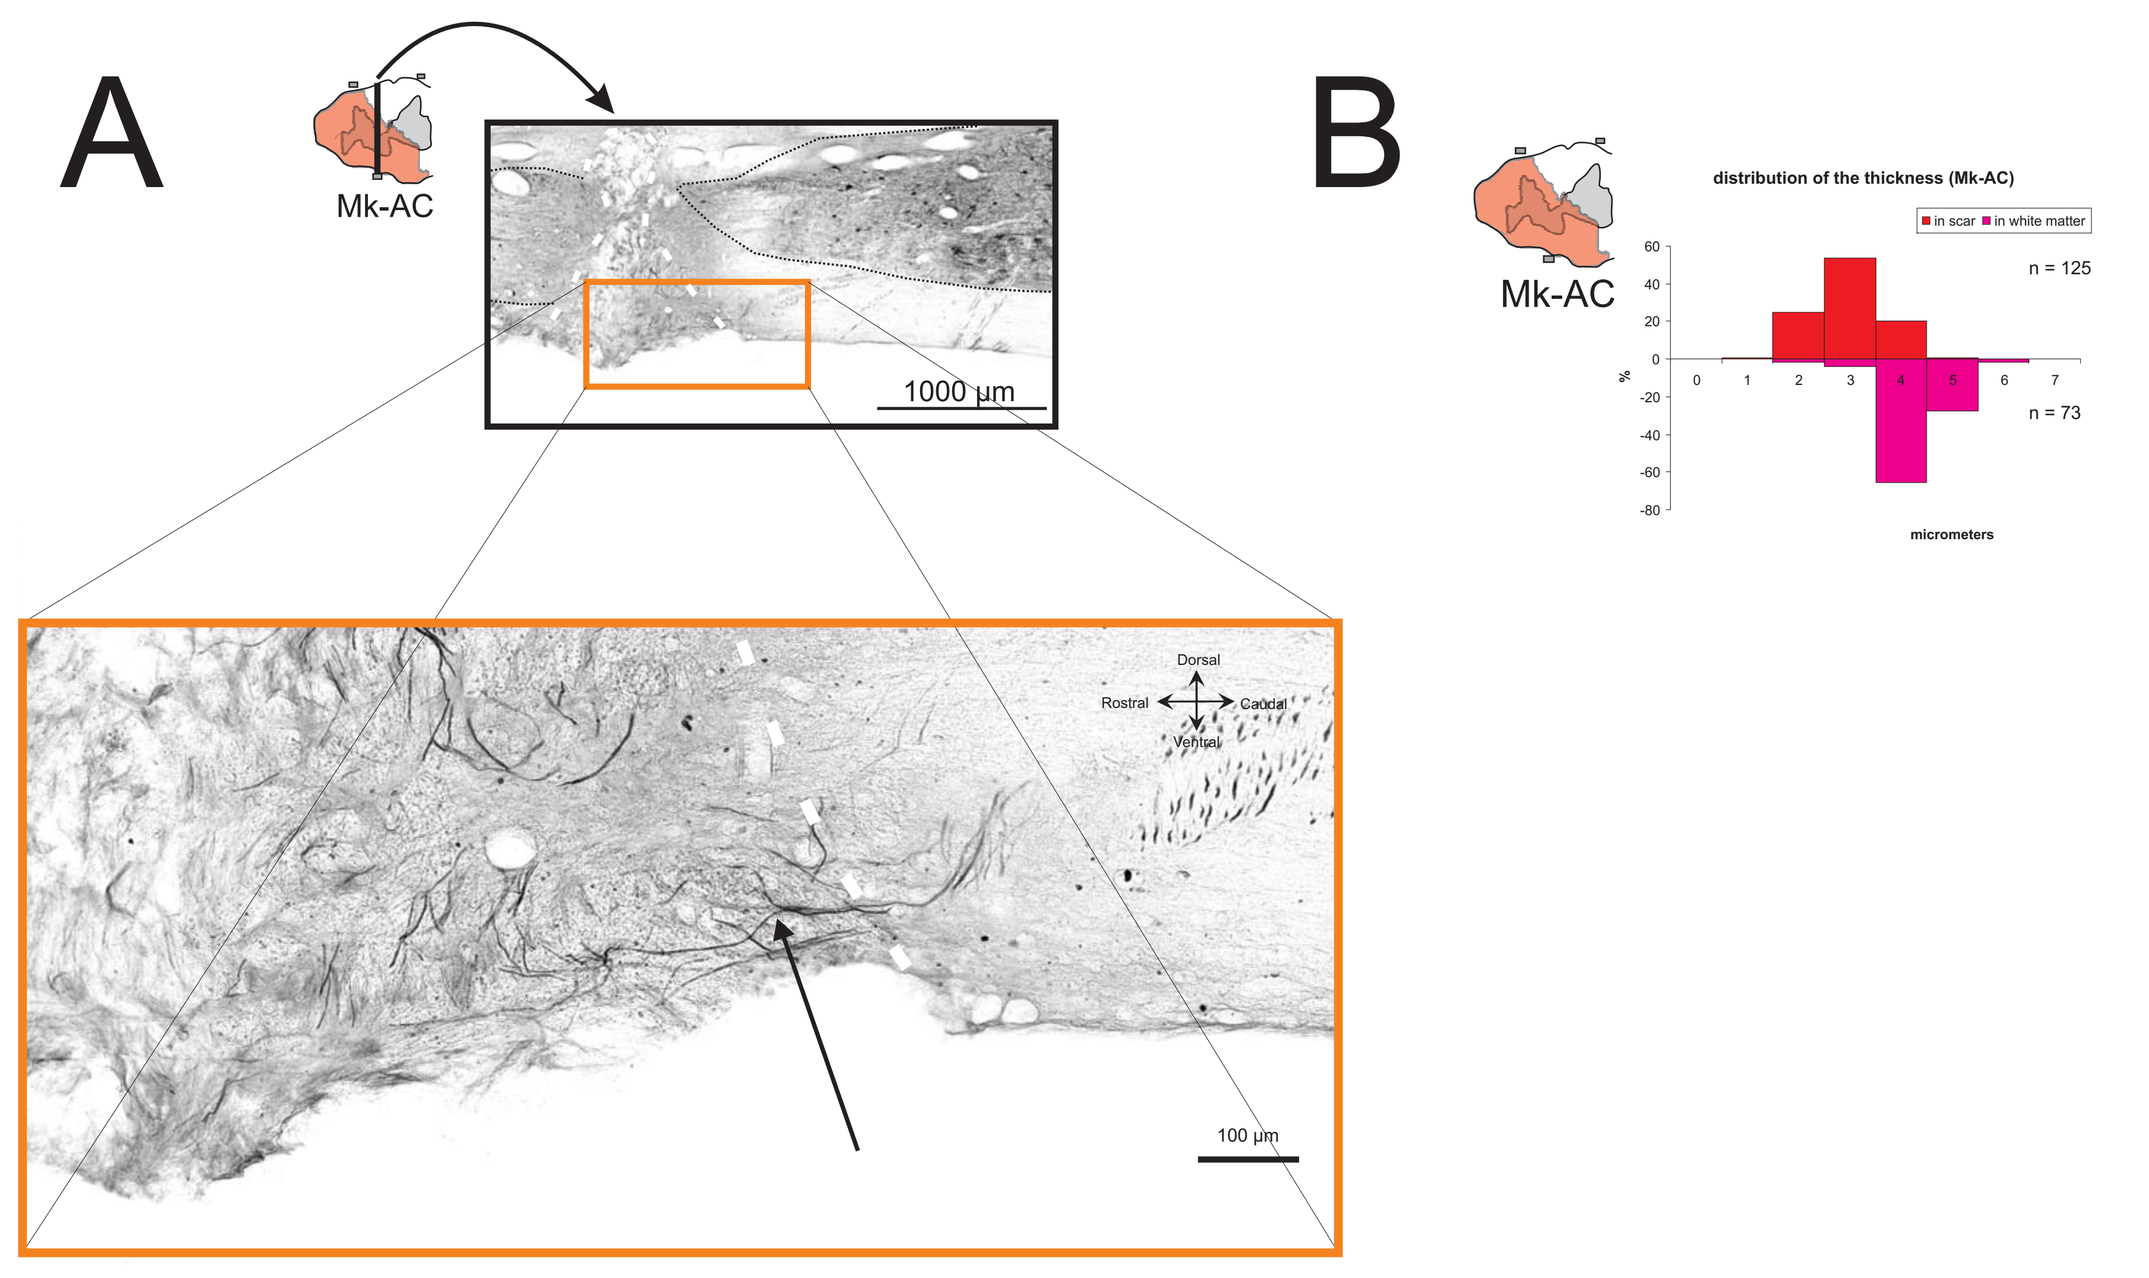

A
Mk-AC
1000 µm
Dorsal
Rostral
Caudal
Ventral
100 µm
B
distribution of the thickness (Mk-AC)
n = 125
n = 73
micrometers

• The diameter of the SMI-32 (+) fibers that belong to the ventral root do not vary as they penetrate into the scar (A) and the fibers width distribution is different in most animal when comparing the scar and the white matter (B).

• This suggests that some of "thin" fibers do not originate from ventral roots.

7) The number of SMI-32 (+) fibers present into the scar is independent of the treatment given.

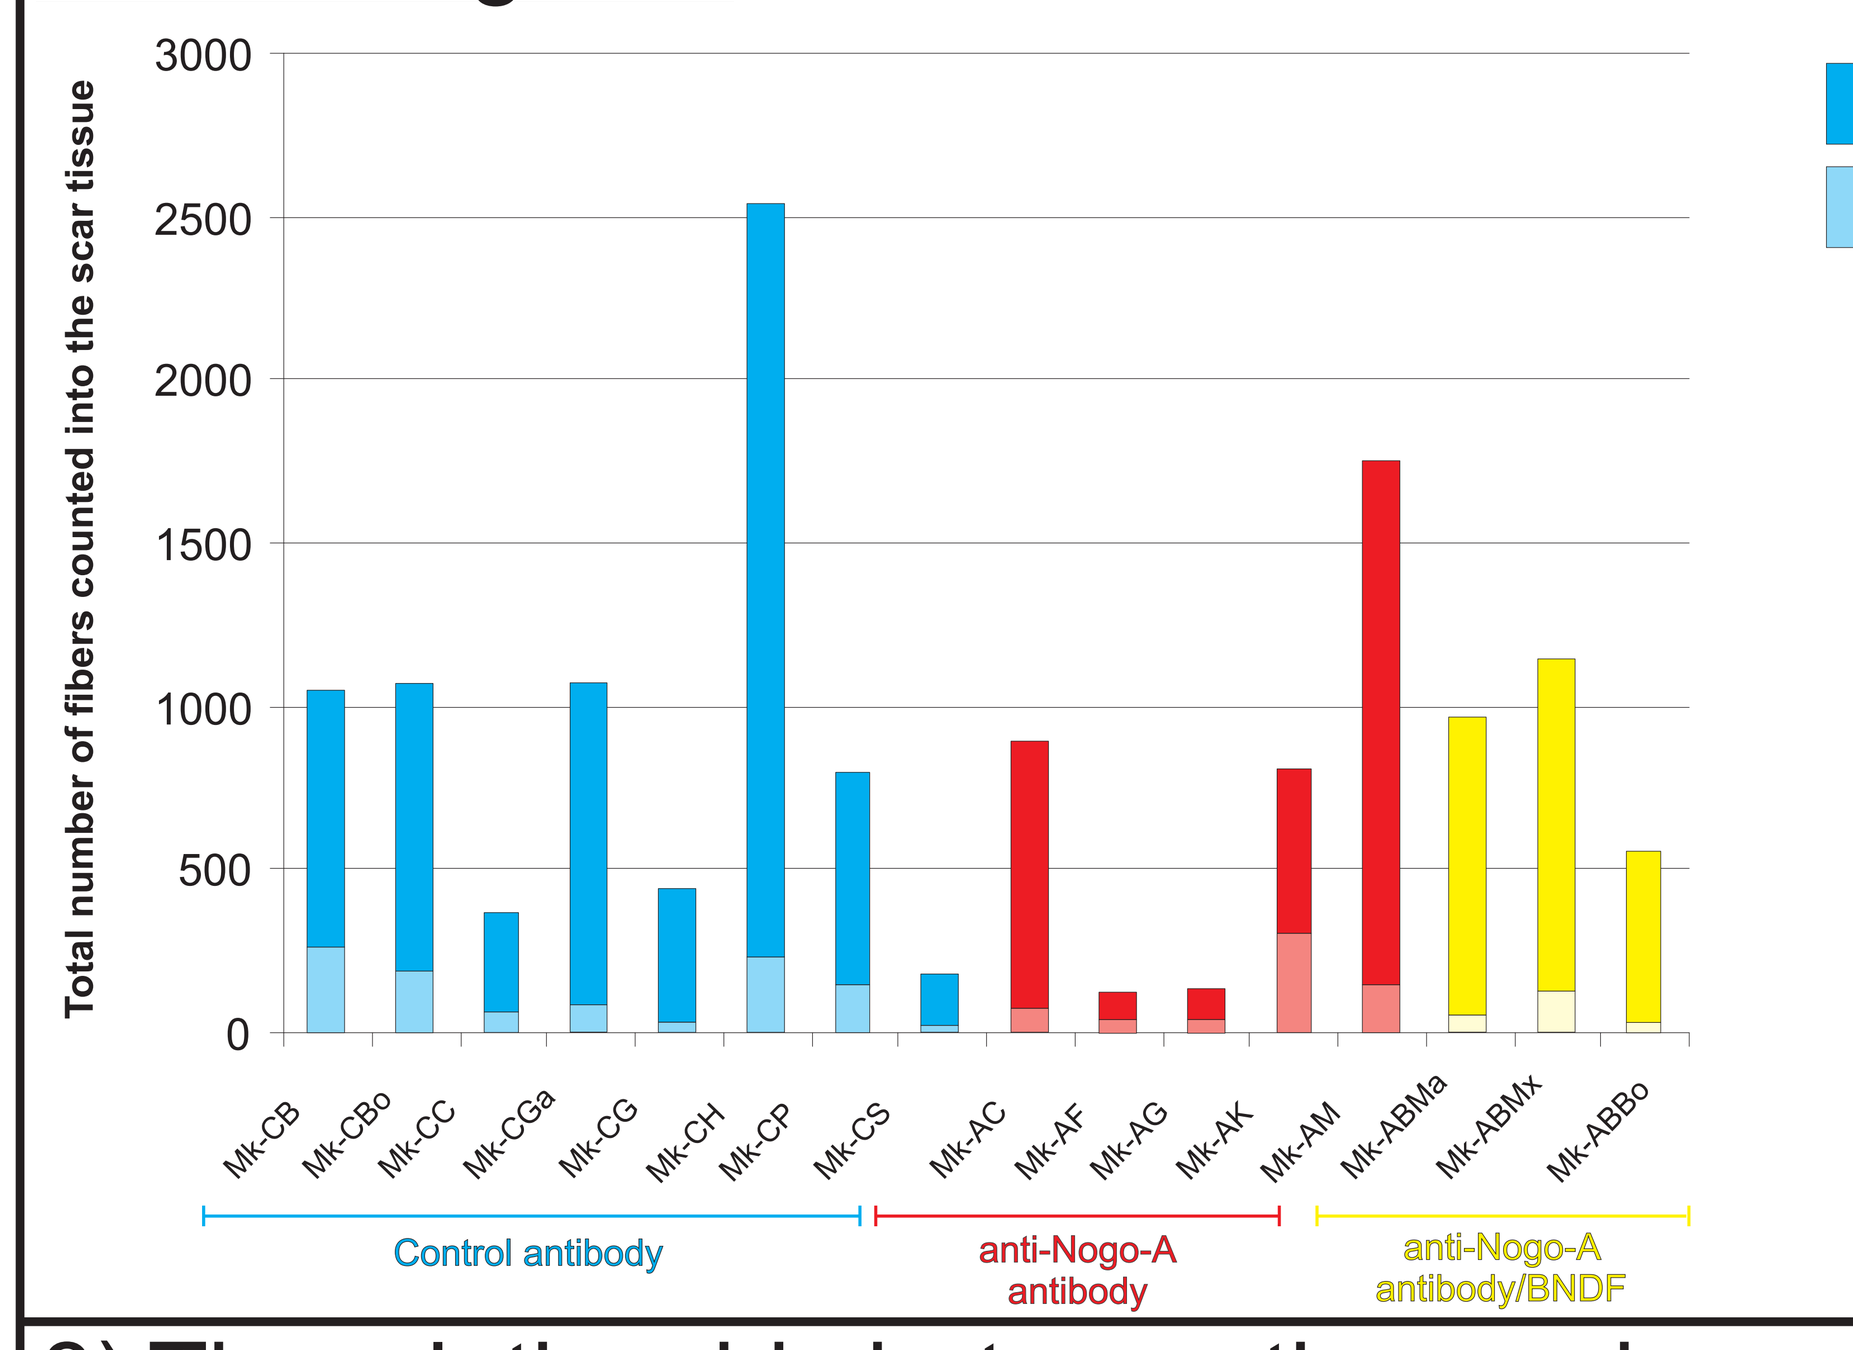

Total number of fibers counted into the scar tissue
Thick Fibers > 3.1 µm
Thin Fibers < 3.1 µm
Mk-CB
Mk-CBo
Mk-CC
Mk-CGa
Mk-CG
Mk-CH
Mk-CP
Mk-CS
Mk-AC
Mk-AF
Mk-AG
Mk-AK
Mk-AM
Mk-ABMa
Mk-ABMx
Mk-ABBo
Control antibody
anti-Nogo-A antibody
anti-Nogo-A antibody/BNDF

• Neither the anti-Nogo-A antibody treatment alone, nor the combined treatment with the neurotrophic factor BDNF did promote the colonization of the scar tissue.

8) The relationship between the number and the cumulated fiber length of SMI-32 (+) fibers.

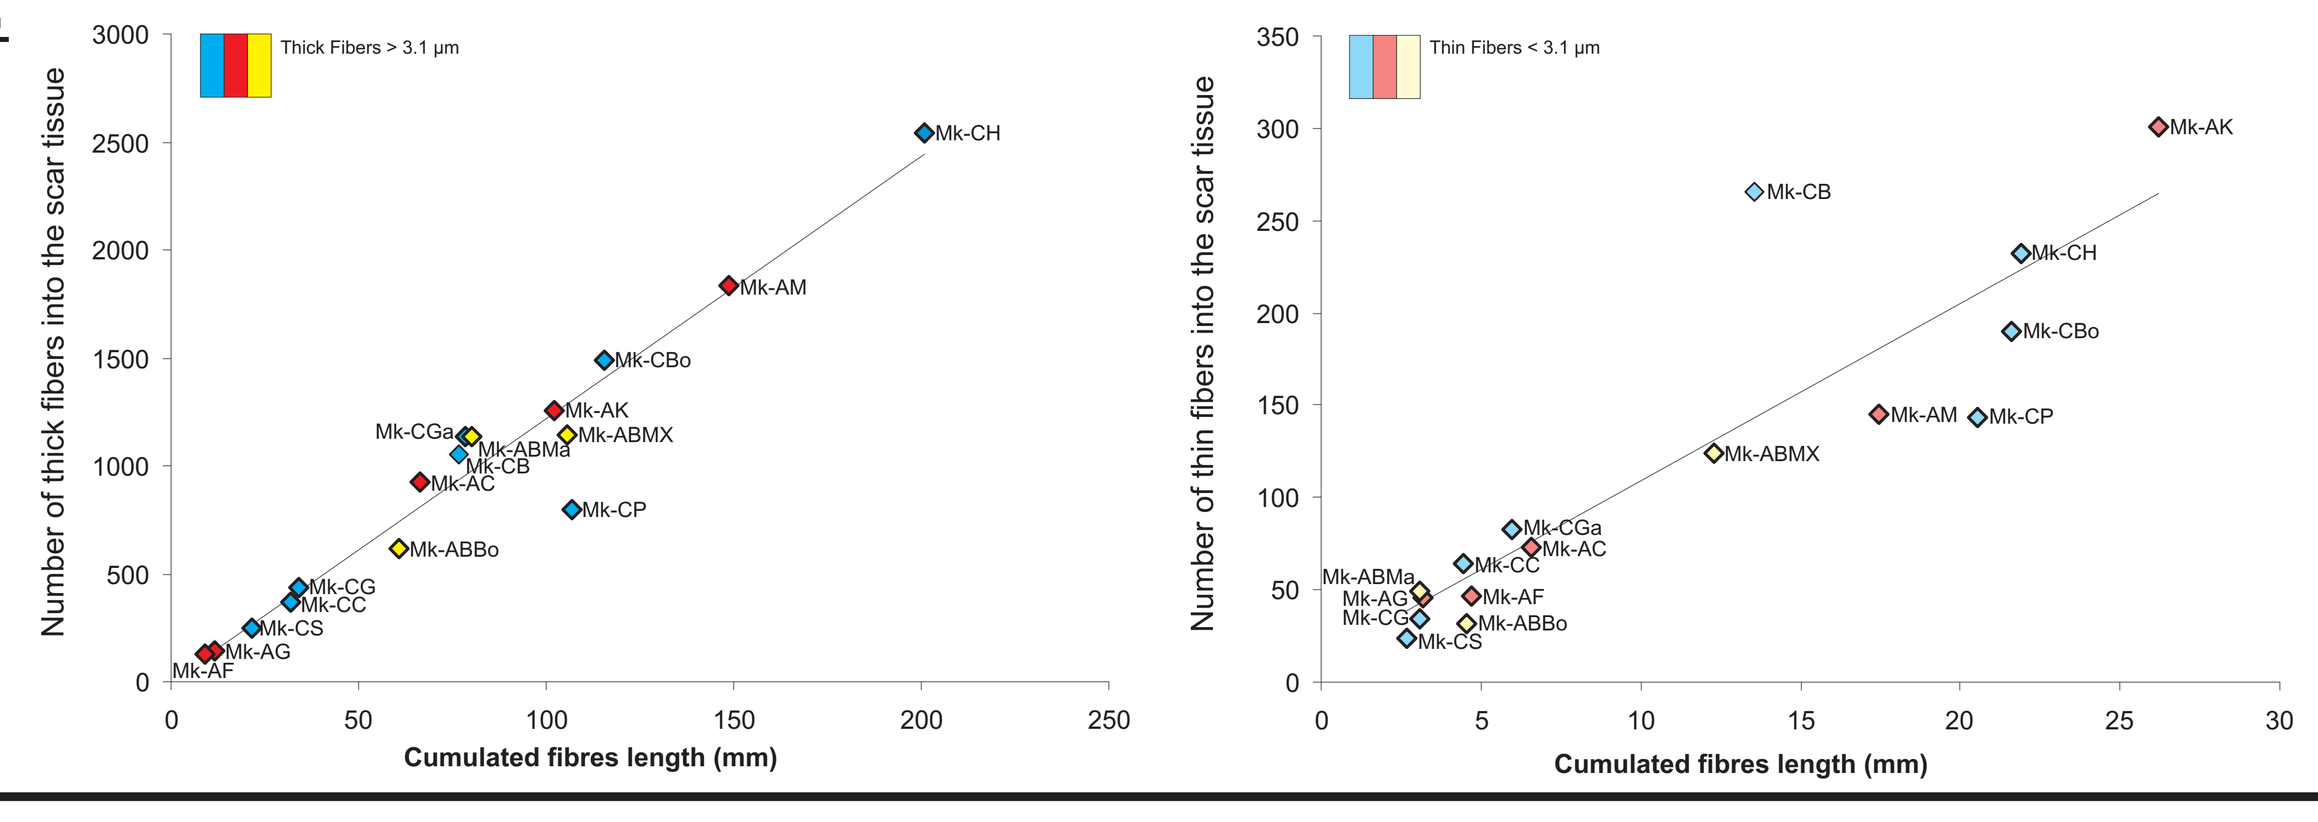

Thick Fibers > 3.1 µm
Number of thick fibers into the scar tissue
Thin Fibers < 3.1 µm
Number of thin fibers into the scar tissue
Cumulated fibres length (mm)

• The correlation between the number and the cumulated fiber lenght is linear and not affected by both growth promoting treatments.

Marie-Laure.Beaud@unifr.ch